Improving Skin Antisepsis: 2% No-Rinse CHG Cloths Improve Antiseptic Persistence on Patient Skin Over 4% CHG Rinse-Off Solution

Marcia Ryder, PhD, MS, RN, Research & Consulting: Medical Biofilm-related Infections and Vascular Access and Jodi Balbinot, BS

OVERVIEW

The Centers for Disease Control (CDC) guidelines for prevention of surgical site infections (SSIs) published a category IB recommendation as follows: "Require patients to shower or bathe with an antiseptic agent on at least the night before the operative day."[1] In order for a product to be classified a skin antiseptic by the FDA, the product must show rapid reduction in resident and transient microbes in the surgical field prior to incision, and maintain effectiveness for a minimum of 6 hours after application.[2]

Currently, there is confounding evidence regarding the benefits of preoperative bathing or showering in relation to prevention of SSIs. It is well known that harmful bacteria, such as methicillin-resistant *Staphylococcus aureus* and vancomycin-resistant *Enterococcus*, attribute to mortality, extended length of stay, and significant costs.[3-6] Chlorhexidine is proven to kill these and other harmful bacteria.[6,7] Chlorhexidine gluconate (CHG) is an FDA-approved skin antiseptic available in a 4% solution that is rinsed off after use in a bath or shower (4% CHG solution) and an alcohol-free 2% CHG impregnated no-rinse cloth (2% CHG cloth). Recommendations, such as the above CDC recommendation,[1] strongly recommend the use of an antiseptic agent at least the night before the operative day to eliminate harmful bacteria; however, a large Cochrane review found the "use of chlorhexidine for preoperative bathing or showering is unlikely to prevent surgical site infection".[8]

The Cochrane review came to this conclusion by evaluating the results of studies which utilized the 4% CHG solution. Residual CHG on the skin after application may influence the efficacy of its use as a preoperative skin antiseptic. This study was designed to compare and evaluate the delivery of the CHG to the skin by the following skin cleansing and preparation products: 4% CHG solution (Hibiclens®, Mölnlycke Health Care US, LLC, Norcross, Georgia 30092) and 2% CHG cloths (Sage® 2% CHG Cloth, Sage Products Inc, Cary, IL; equivalent to 500 mg chlorhexidine gluconate per cloth).

REFERENCES

1 Nichols RE. Preventing Surgical Site Infections: A Surgeon's Perspective. *Emerging Infectious Diseases.* 2001;7(2):220-224. Available at: http://www.cdc.gov. Accessed May 31, 2007

2 Topical Antimicrobial Drug Products for Over-the-Counter Human Use: Tentative Final Monograph for Health-Care Antiseptic Drug Products. 1994. Available at: http://www.fda.gov/cder/otcmonographs/antimicrobial/antimicrobial_antiseptic_TF_PR_19940617.pdf. Accessed on June 16, 2007. (No authors available on the website).

3 Stone PW, Braccia D, Larson E. Systematic review of economic analyses of health care-associated infections. *Am J Infect Control.* 2005;33:501-509.

4 Kirkland KB, Briggs JP, Trivette SL, et al. The impact of surgical-site infections in the 1990s: attributable mortality, excess length of hospitalization, and extra costs. *Infect Control Hosp Epidemiol.* 1999;20:725-730.

5 Engemann JJ, Carmeli Y, Cosgrove SE, et al.Adverse clinical and economic outcomes attributable to methicillin resistance among patients with Staphylococcus aureus surgical site infection. *Clin Infect Dis.* 2003;36(5):592-598.

6 Edmiston C, Seabrook GR, Johnson CP, et al. Comparison of a New and Innovative 2% Chlorhexidine Gluconate (CHG) Impregnated Preparation Cloth with the Standard 4% CHG Surgical Skin Preparation. Poster presented at the 2007 Association of periOperative Registered Nurses (AORN) Congress, Orlando, FL Mar 2007.

7 DeBaun B. New Alcohol-Free 2% CHG Solution Reduced Bacterial Counts of Drug-Resistant Acinetobacter and MRSA by 99.9%. Poster presented at the 2007 Association of periOperative Registered Nurses (AORN) Congress, Mar 2007.

8 Webster J, Osborne S. Preoperative bathing or showering with skin antiseptics to prevent surgical site infection. Art. No.: CD004985. DOI: 10.1002/14651858.CD004985.pub3. Available at: http://www.cochrane.org/reviews/en/ab004985.html. Accessed May 31, 2007.

METHODS

This was a prospective, randomized study with a total cohort of 24 subjects (4 males, 20 females; median age of 39, ranging from 30 to 58 years). All subjects used both products to eliminate any variation that may be caused by skin type, showering technique, etc. between subjects. The study was comprised of two groups: Groups A (n=12) and B (n=12).

CHG Prep and Test Protocol:

- **Test Day 1**:* Group A showered **morning of** test with **4% rinse-off CHG**
 Group B showered the **night before** and **morning of** test with **4% rinse-off CHG**
- **Test Day 8**:** Group A wiped down with **no-rinse 2% CHG cloths** the **morning of** test
 Group B wiped down with **no-rinse 2% CHG cloths** the **night before** and **morning of** test

* subjects applied 4% CHG solution to entire body only from neck down (instructed not to use near eyes or ears); instructed to use as much 4% CHG solution as needed to wash body; used one towel for drying hair; used another towel to pat body dry; instructed not to use any lotions, moisturizers, or perfumes.

** subjects used one cloth for each of the following areas (wiping each area for ~30 seconds each): neck, chest, and abdomen; arms; one leg and foot; other leg and foot; buttocks and genitals; back; allowed each area to dry for one minute, did not rinse; instructed not to use any lotions, moisturizers, or perfumes.

CHG Prepping Protocol

4% CHG rinse-off solution:

On Day 1, both groups showered normally with a standard washcloth and 4% CHG rinse-off solution provided; subjects were instructed not to use regular soap after 4% CHG solution was used.

2% CHG no-rinse cloths:

On Day 8, both groups did not shower, but instead wiped down entire body with three packages (6 cloths total) of the 2% CHG no-rinse cloths provided. Subjects used 1 cloth to wipe each of the following areas for ~30 seconds each: neck, chest and abdomen; arms; left leg and foot; right leg and foot; buttocks and genitals; and back. Each area was allowed to dry for 1 minute, and subjects did not rinse.

Wiping with 2% no-rinse CHG cloth

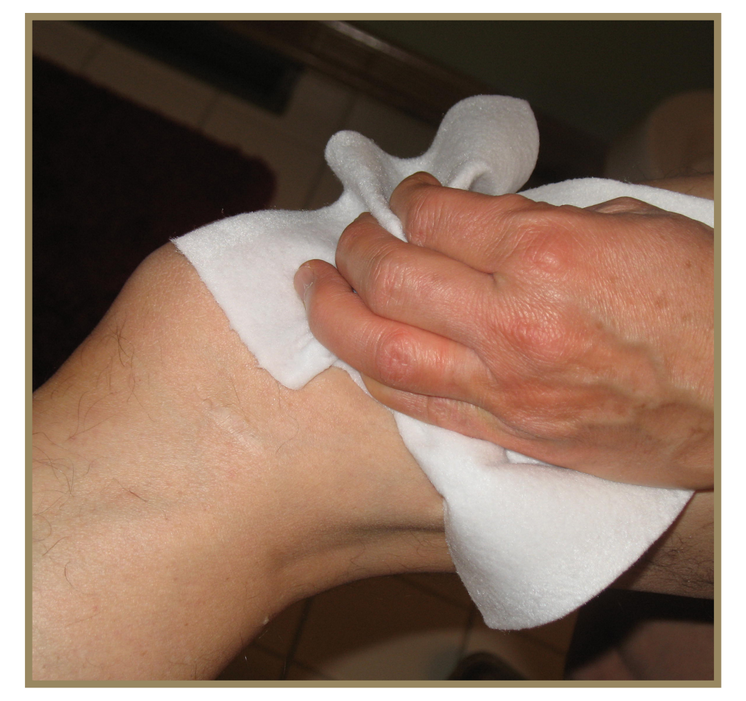

Washout period:

All subjects returned to normal showering routine during the washout period between Days 1 and 8, to allow for removal of any residual CHG from Day 1.

CHG Testing

At the time of testing, the 2% CHG cloths and the 4% CHG solution bottles were weighed to determine how much product had been used. Sampling for CHG residual was performed 3 and 10 hours after washing in Group A (morning prep). Sampling for Group B (night before and morning prep) was done 3 hours after the morning wash. A sterile swab was premoistened with 105 µl of sterile water and used to sample the skin with circular motion ~1" in diameter for ~10 seconds. Sampling sites included the abdomen, behind each knee, and the left and right forearms.

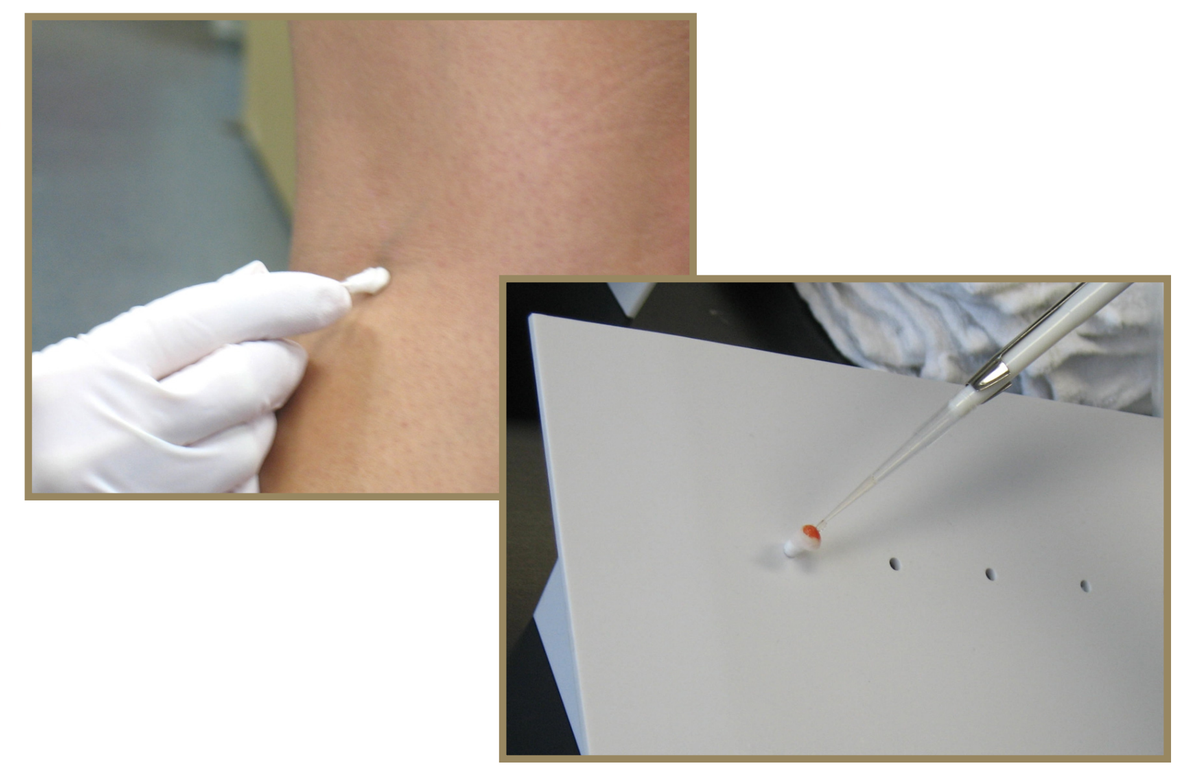

Cetyltrimethylammonium bromide and sodium hypobromite were used as the reagents to create a color response to determine the amount of CHG residual. The test swabs were compared to swabs inoculated with known amounts of CHG (16µg-2100µg) and tested with reagent.

RESULTS

Is there a correlation between the amount of CHG residual on the skin after use of the 4% CHG solution compared to the 2% CHG cloth?

There was no correlation between the amount of 4% CHG solution used and the residual amount left on the skin (correlation coefficient 0.22, P=0.29). However, when the same analysis was done for the 2% CHG cloth, there was a significant correlation between the amount of product used and the amount of residual on the skin (correlation coefficient 0.68, P=0.0003).

Is there a difference in the amount of 4% CHG solution used compared to the amount of CHG used with the 2% CHG cloth?

The amount of solution used during showering with 4% CHG solution and wiping with the 2% CHG cloth was comparable. There was no statistical difference in the amount of CHG solution used between Day 1 (4% CHG solution) and Day 8 (2% CHG cloth; P=0.63).

Quantification of CHG

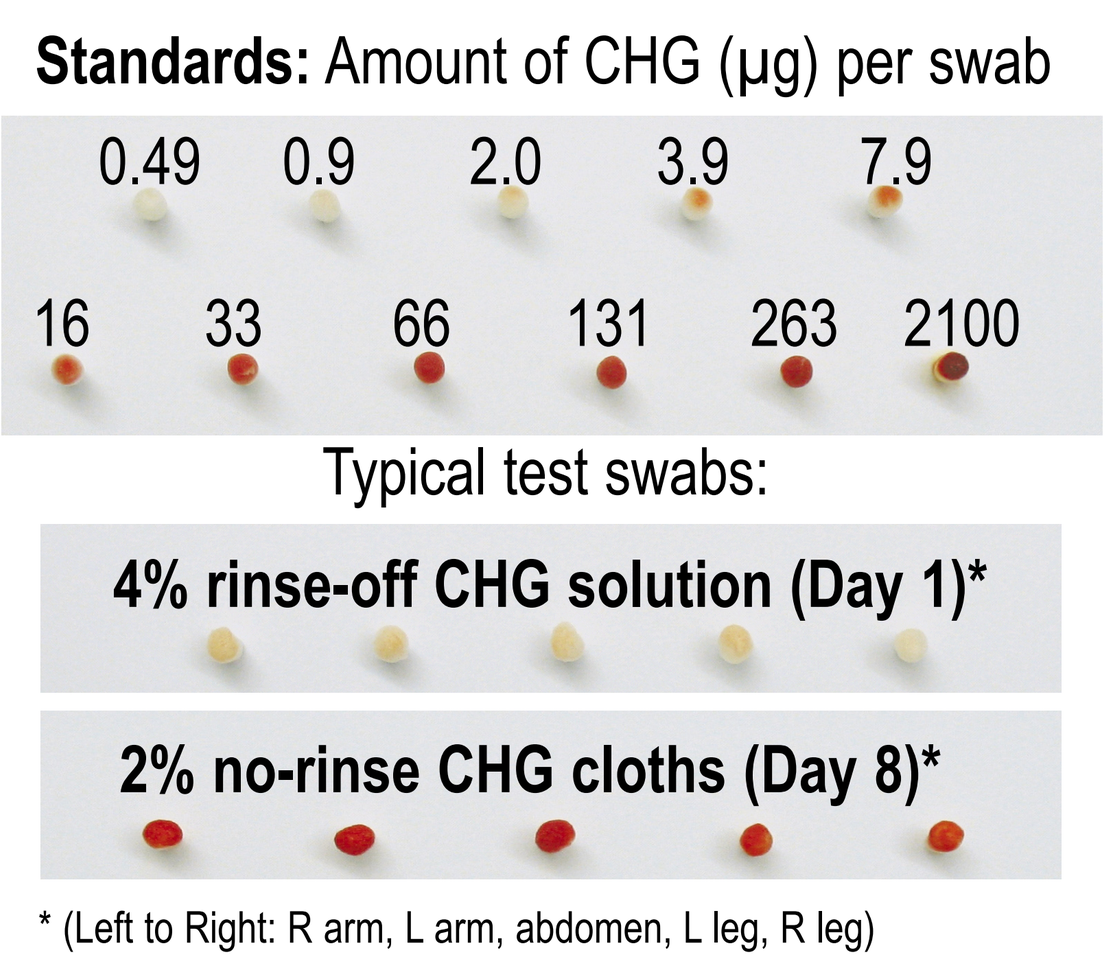

Standards: Amount of CHG (µg) per swab
0.49 0.9 2.0 3.9 7.9
16 33 66 131 263 2100
Typical test swabs:
4% rinse-off CHG solution (Day 1)*
2% no-rinse CHG cloths (Day 8)*

* (Left to Right: R arm, L arm, abdomen, L leg, R leg)

Is there a difference in the residual CHG left on the skin when the 4% CHG solution is used compared to the use of the 2% CHG cloth?

In both groups, the 2% CHG cloth subjects had more residual CHG on their skin than the 4% CHG solution subjects.

Is there a difference in the residual CHG left on the skin after one or two preps with the 4% CHG solution compared to the 2% CHG cloth?

Two preps with 4% CHG solution showed no more residual CHG than one prep (P=0.137).

Two preps with the 2% CHG cloth showed more residual CHG than one prep (P=0.016).

Residual CHG: Prepping Once vs Prepping Twice

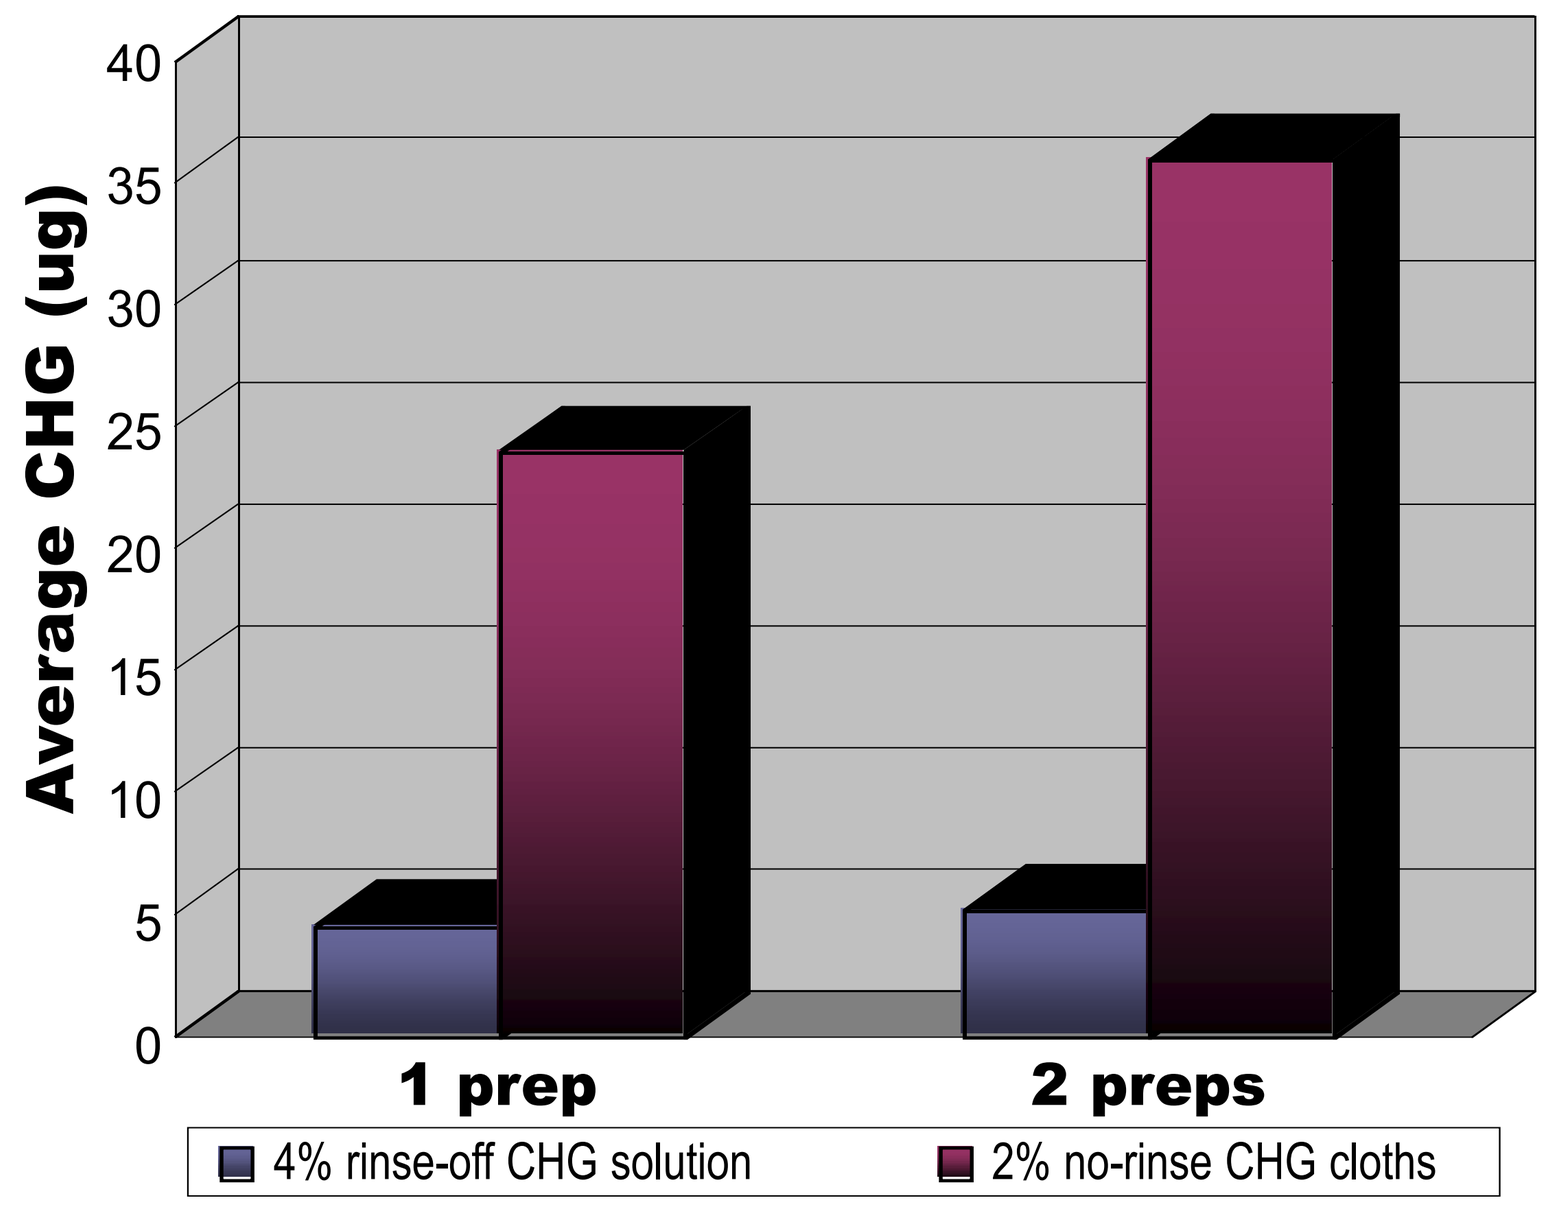

Average CHG (ug)
0
5
10
15
20
25
30
35
40
1 prep
2 preps
4% rinse-off CHG solution
2% no-rinse CHG cloths

Is there a difference in the CHG residual left on the skin at three and ten hours after the use of the 4% CHG solution compared to the 2% CHG cloth?

Group A subjects were tested on Day 1 and Day 8 at three hours and ten hours post-prepping. There was no change in the amount of CHG detected on the skin at three hours and ten hours for 4% CHG solution (P=0.47) or the 2% CHG cloths (P=0.16).

Is there a difference in the amount of CHG residual left on the skin at each body sample site within each test group (4% CHG solution and 2% CHG cloth)?

In both groups, there was a significant difference in the amount of residual CHG detected between different sample sites (P=0.003).

Average Residual CHG by Sample Area

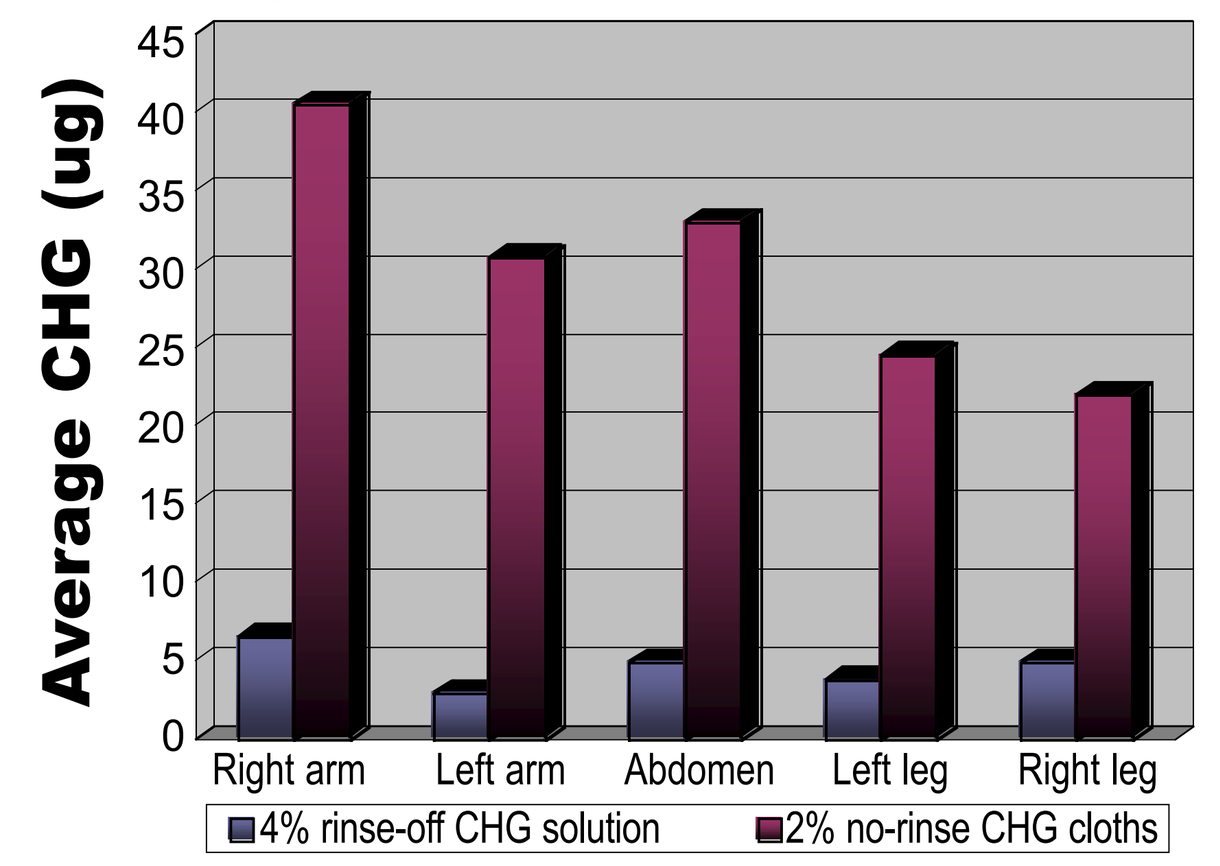

Average CHG (ug)
0
5
10
15
20
25
30
35
40
45
Right arm
Left arm
Abdomen
Left leg
Right leg
4% rinse-off CHG solution
2% no-rinse CHG cloths

Is there a difference in the amount of CHG residual left on the skin at each body sample site between each test group (4% CHG solution and 2% CHG cloth)?

When each individually swabbed body area (sampling site) was compared between Day 1 and Day 8, there was more CHG residual for each area tested after prepping with 2% CHG cloths than with the 4% CHG solution.

CONCLUSIONS

This study was done to determine if there is a difference in the amount of residual CHG left on the skin when prepping with a 4% rinse-off application of CHG compared to that of a 2% no-rinse application.

The amount of CHG that remains on the skin after a no-rinse application is significantly higher than a CHG application that is rinsed off.

This is true despite the fact that the rinse-off application has a higher concentration of CHG (4%) than the no-rinse cloth (2%).

No correlation was found between the amount of rinse-off product used and the residual on the skin.

This finding indicates that most of the CHG is likely rinsed off the skin during or after the application of the product, leaving very little CHG on the skin. This could contribute to the efficacy of the no-rinse CHG cloths as compared to the rinse-off product.

A higher residual CHG quantity is attained by prepping twice with the 2% CHG cloth, whereas the 4% CHG solution shows no additional residual quantity after the second prep.

Difference in residual CHG between sample sites may reflect more difficulty in prepping certain body parts.

The significant difference of the amount of residual CHG detected when comparing sample sites within the groups reflects the difficulty patients may have in prepping certain body areas, such as behind the knee. Therefore, especially for total knee patients, instructions to the patient need to highlight the importance of paying special attention to this area during pre-surgery skin preparation.

20983